Predictors and prognostic implication of pathologic fracture, spinal cord compression and bone surgery following diagnosis of metastatic prostate cancer

E Onukwugha[1], C Yong[1], CD Mullins[1], B Seal[2], A Hussain[3]

[1] University of Maryland School of Pharmacy, Baltimore, MD

[2] Bayer HealthCare Pharmaceuticals, Inc.

[3] University of Maryland School of Medicine, Baltimore, MD

Background

- Skeletal-related events (SREs) are associated with an increased hazard of prostate cancer-specific mortality among men diagnosed with prostate cancer (PCa) and bone metastasis (BM).
- Information also is limited regarding the separate roles of components of SREs including pathological fracture (PF), spinal cord compression (SCC), and bone surgery (BS).

Objective

To estimate the hazard of PCa-specific and all-cause mortality associated with SREs, as a cluster of events as well as by component.

Methods

- We analyzed PCa-specific and all-cause mortality among elderly SEER-Medicare stage IV PCa patients diagnosed between 2000 and 2007.

- Treatment-related data from 1999 to 2009 were extracted from linked Medicare claims files.

- Patients were censored if they enrolled in an HMO or lost Part A and/or B enrollment or if they were alive at the end of the period (December, 2009)

Study inclusion and *exclusion* criteria:
- Continuous enrollment in Medicare Part A and B during the 13 months prior to and including the month of diagnosis
- Diagnosis of distant metastasis (i.e. M1 PCa)

- *Health maintenance organization (HMO) enrollment during the 12 months prior to and including the month of diagnosis;*
- *History of other cancers within 5 years prior to PCa diagnosis;*
- *PCa diagnosis occurred at the autopsy*

Measures:
- Occurrence of PF, SCC, and BS following PCa diagnosis was identified using Medicare claims and utilized in defining an SRE using three approaches.
 - Measure 1: SRE concurrent with or following a claim with a BM ICD 9 diagnosis code
 - Measure 2: SRE claim includes BM ICD 9 coding directly on the SRE claim
 - Measure 3: SRE is not anchored to a claim with a BM ICD 9 code.

Statistical analysis:
- Statistical analyses examined the association between SREs and PCa-specific mortality using the Cox proportional hazards regression model in the original sample and in a propensity score-matched sample (PSMS)
- Stratified direct-adjusted survival curves were produced in the case of statistically significant non proportional hazards associated with an SRE.

Results

- Application of inclusion criteria resulted in 7,062 patients (1,776 in PSMS).
- PCa-specific and all-cause mortality were 54% and 80% at a median (mean; min; max) follow up of 609 days (837; 30; 3,653).
- The average age for the sample was 78 years and 14% were non-Hispanic African American.
- The proportion with any event, SCC, PF, and BS differed according to the measure utilized (Table 1).

Table 1. Descriptive statistics for individuals diagnosed with stage IV M1 prostate cancer, stratified by disease-specific mortality (N=7,062)

Variable	Full sample N=7,062 % (N)	PCa-specific mortality No N=3,279 %	Yes N=3,783 %	p-value
SRE concurrent with or following a claim with a BM ICD 9 diagnosis code				
Skeletal-related event (SRE)	12.6% (N=892)	9.8%	15.1%	<0.0001
Spinal cord compression (SCC)	7.4% (N=524)	5.7%	8.9%	<0.0001
Pathological fracture (PF)	4.3% (N=306)	3.1%	5.4%	<0.0001
Bone surgery (BS)	1.9% (N=133)	2.0%	1.8%	0.69
Bone metastasis ICD 9 code on SRE claim				
Skeletal-related event (with BM code)	9.7% (N=682)	7.4%	11.6%	<.0001
Spinal cord compression (with BM code)	5.5% (N=385)	4.2%	6.6%	<.0001
Pathologic fracture (with BM code)	4.0% (N=284)	3.0%	4.9%	<.0001
Bone surgery (with BM code)	1.2% (N=83)	1.1%	1.2%	0.733
Any SRE post diagnosis of M1 prostate cancer				
Skeletal-related event	17.1% (N=1,205)	15.4%	18.5%	<0.001
Spinal cord compression	10.1% (N=712)	8.9%	11.1%	0.003
Pathologic fracture	5.4% (N=381)	4.2%	6.4%	<.0001
Bone surgery	3.1% (N=219)	3.8%	2.5%	0.003
SRE (Measure 1) disaggregated				
Spinal cord compression (w/o surgery)	6.6% (N=465)	4.8%	8.1%	0.0006
Spinal cord compression (with surgery*)	0.8% (N=59)	0.9%	0.8%	0.67
Pathologic fracture (w/o surgery)	2.3% (N=162)	1.7%	2.8%	0.002
Pathologic fracture (with surgery*)	2% (N=144)	1.4%	2.6%	0.0004
Bone surgery only	1.9% (N=133)	2.0%	1.8%	0.69

*Concurrent bone surgery

Table 2: Covariate-adjusted hazard ratios for skeletal-related events (SREs) from Cox proportional hazards models for prostate cancer–specific mortality, by type of SRE**

Variables	HR	95 % CI	p-value
SRE concurrent with or following a claim with a BM ICD 9 diagnosis code			
Any SRE	1.27	1.16 - 1.39	<0.001
Spinal cord compression (SCC)	1.39	1.24 - 1.55	<0.001
Pathological fracture (PF)	1.19	1.03 - 1.37	0.01
Bone surgery (BS)	0.76	0.60 - 0.97	0.03
Bone metastasis ICD 9 code on SRE claim			
Any SRE	1.31	1.18 - 1.45	<0.0001
SCC	1.37	1.20 - 1.56	<0.0001
PF	1.17	1.01 - 1.37	0.04
BS	0.86	0.64 - 1.15	0.31
Any SRE post diagnosis of M1 prostate cancer			
Any SRE	1.07	0.98 - 1.16	0.13
SCC	1.18	1.06 - 1.30	0.002
PF	1.15	1.004 - 1.31	0.044
BS	0.59	0.48 - 0.72	<0.001

*Mortality models included patient demographic and clinical factors as well as registry and diagnosis year fixed effects.
M1: metastatic; PCa: prostate cancer

Results

Table 3: All-cause and non prostate cancer-specific mortality models

Variables	All-cause mortality HR	CI	p-value	Non PCa-specific mortality HR	CI	p-value
SRE concurrent with or following a claim with a BM ICD 9 diagnosis code						
Any SRE	1.14	1.06 - 1.24	<.0001	0.87	0.75 – 1.00	0.0503
Bone metastasis ICD 9 code on SRE claim						
Any SRE	1.21	1.11 - 1.31	<.0001	0.95	0.81 - 1.12	0.533
Any SRE post diagnosis of M1 prostate cancer						
Any SRE	1.05	0.98 - 1.12	0.205	0.97	0.86 - 1.09	0.619

SRE: skeletal related event
PCa: prostate cancer

- The hazard of PCa-specific mortality associated with an SRE varied according to the measure utilized (Table 2).

- The hazard of all-cause mortality also varied with the measure utilized and results were similar to results from the PCa-specific mortality model (Table 3).

- Using Measures 1 and 2 there was a statistically significant interaction with time, indicating that the mortality hazard associated with an SRE increased over time (Figure 1).

- Regarding the SRE components, the HRs on SCC and PF were statistically significantly associated with PCa-specific mortality across the three measures. The results for BS were sensitive to the approach used in defining the event.

- Results for SCC and BS were unchanged from Measure 1 when using a propensity score matched sample.

Figure 1: Direct-adjusted curve for SRE following BM-related claim in M1 PCa sample using Measure 1 (top), Measure 2 (middle), and Measure 3 (bottom)

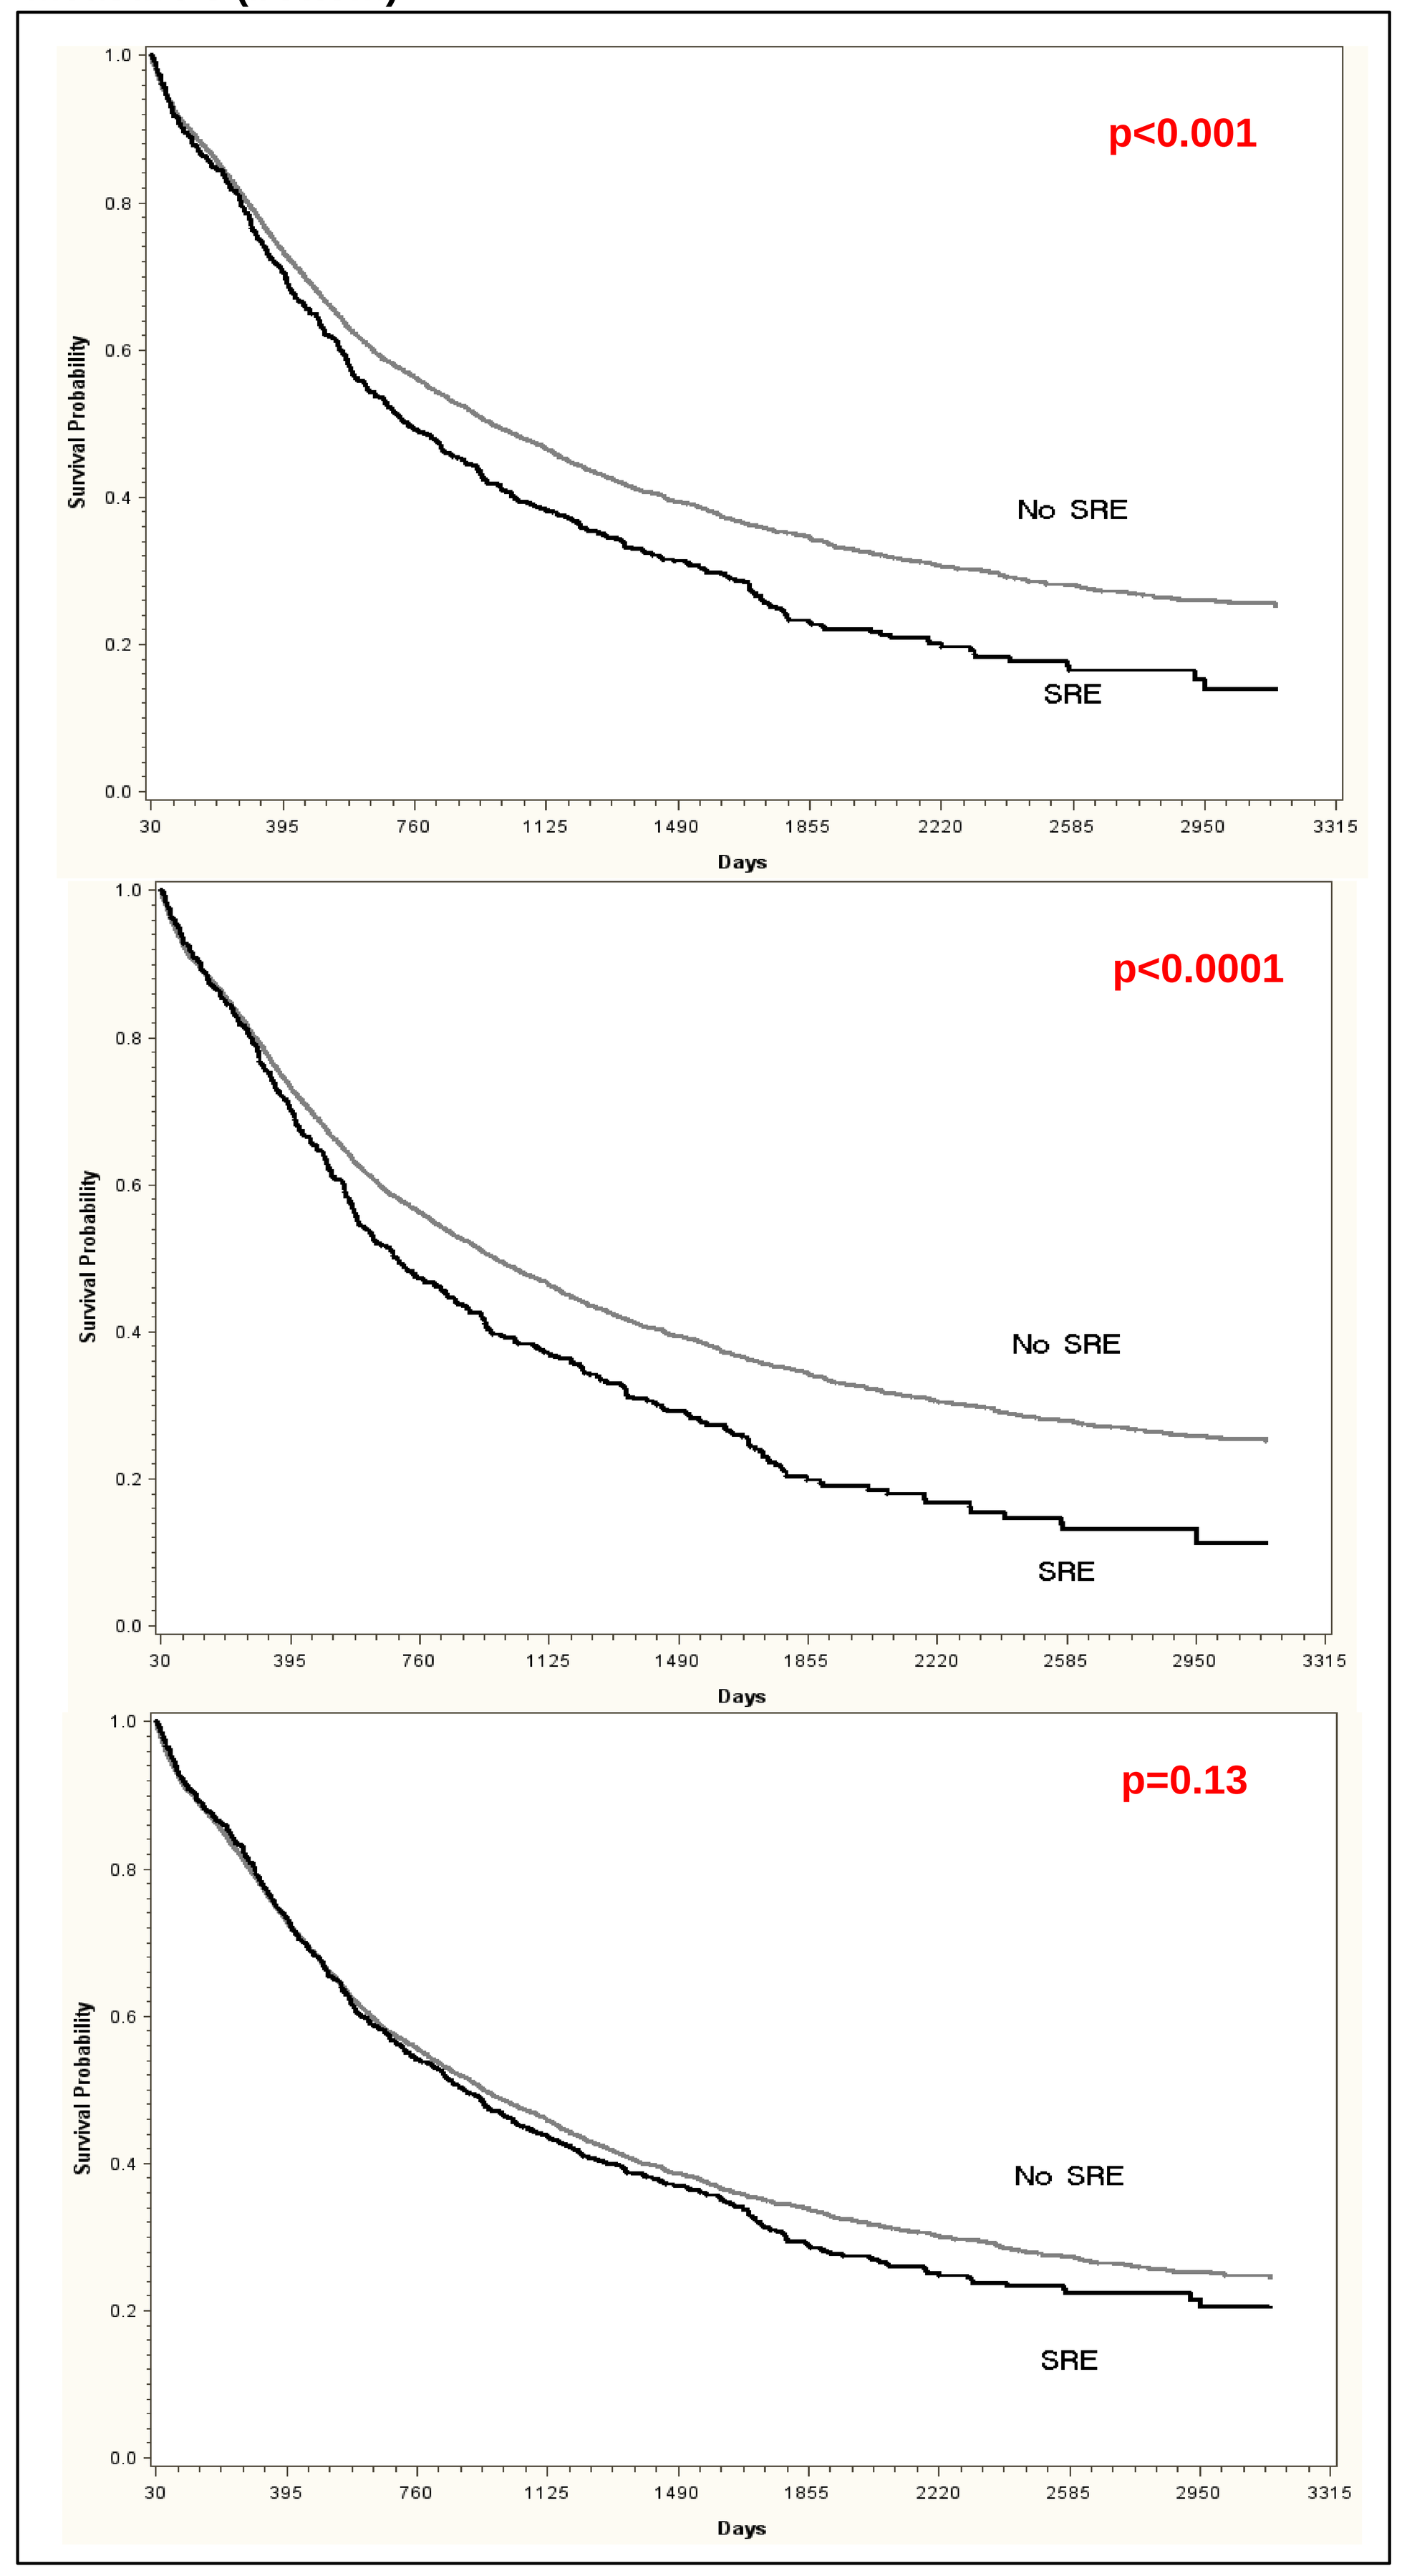
p<0.001
No SRE
SRE
p<0.0001
No SRE
SRE
p=0.13
No SRE
SRE
Survival Probability
Days

Conclusion

Compared to fractures and spinal cord compression, the relationship between bone surgery and PCa-specific mortality is more favorable.

Acknowledgements

Funding for the study was provided by Bayer Healthcare Pharmaceuticals. The authors wish to thank Corinne Woods and Lori Walker from the University of Maryland School of Pharmacy's Pharmaceutical Research Computing for programming assistance on the primary datasets.

Contact Information

Eberechukwu Onukwugha, PhD
Department of Pharmaceutical Health Services Research
University of Maryland School of Pharmacy
eonukwug@rx.umaryland.edu